Updated Survival and Safety Data From RADIANT-3: A Randomized, Double-Blind, Placebo-Controlled, Multicenter, Phase III Trial of Everolimus in Patients With Advanced Pancreatic Neuroendocrine Tumors

Catherine Lombard-Bohas,[1] Eric Van Cutsem,[2] Jaume Capdevila,[3] Elisabeth G. E. de Vries,[4] Paola Tomassetti,[5] Jeremie Lincy,[6] Timothy Hobday,[7] Rodney Pommier,[8] James C. Yao[9]

[1]Hôpital E. Herriot, Hospices Civils de Lyon, France; [2]University Hospital Gasthuisberg/Leuven, Leuven, Belgium; [3]Vall d'Hebron University Hospital, Barcelona, Spain; [4]University Medical Center, Gröningen, The Netherlands; [5]University Hospital St. Orsola, Bologna, Italy; [6]Novartis Oncology, Basel, Switzerland; [7]Mayo Clinic, Rochester, Minnesota, USA; [8]Oregon Health & Science University, Portland, Oregon, USA; [9]The University of Texas MD Anderson Cancer Center, Houston, Texas, USA

BACKGROUND

- Pancreatic neuroendocrine tumors (pNET) account for approximately 1.3% of the incidence of all pancreatic cancers and 4% to 7% of the incidence of all NET, although both incidences are increasing[1-4]
- At diagnosis, nearly 65% of patients with pNET have advanced metastatic disease, which is associated with a median survival time of 24 months[4]
- Although the molecular pathogenesis of sporadic pNET is unknown, data suggest that the phosphatidylinositol 3-kinase/Akt/mammalian target of rapamycin (mTOR) pathway, which is important for the regulation of cell growth, proliferation, and angiogenesis, is involved[5-8]
- Everolimus, an oral mTOR inhibitor, has shown promising antitumor activity in a phase II study in patients with pNET[9]
- Everolimus was approved by the US Food and Drug Administration in May 2011 and was given a positive opinion by the European Union Committee for Medicinal Products for Human Use in July 2011 for the management of progressive pNET that is unresectable, locally advanced, or metastatic
- In the large, prospective, international, multicenter, randomized, double-blind, placebo-controlled, phase III RADIANT-3 trial involving 410 patients with progressing low- or intermediate-grade advanced pNET, everolimus 10 mg/d + best supportive care (BSC) compared with placebo + BSC demonstrated a statistically significant and clinically meaningful 6.4-month prolongation of median progression-free survival (PFS), as assessed by the local investigator (11.0 months vs 4.6 months; hazard ratio [HR], 0.35; 95% confidence interval [CI], 0.27-0.45; *P* < 0.0001)[10]
 - — The benefit provided by everolimus was durable, as evident by the 34% of patients who remained free of disease progression at 18 months[10]

OBJECTIVE

- To evaluate an updated overall survival (OS) analysis (cutoff date February 23, 2011) and the 90-day safety update (cutoff date June 3, 2010) of everolimus + BSC versus placebo + BSC in patients who participated in the RADIANT-3 trial

METHODS

Study Design and Patient Population

- RADIANT-3 was an international, multicenter, double-blind, placebo-controlled, phase III study (**Figure 1**)

Figure 1. Phase III RADIANT-3 study design.

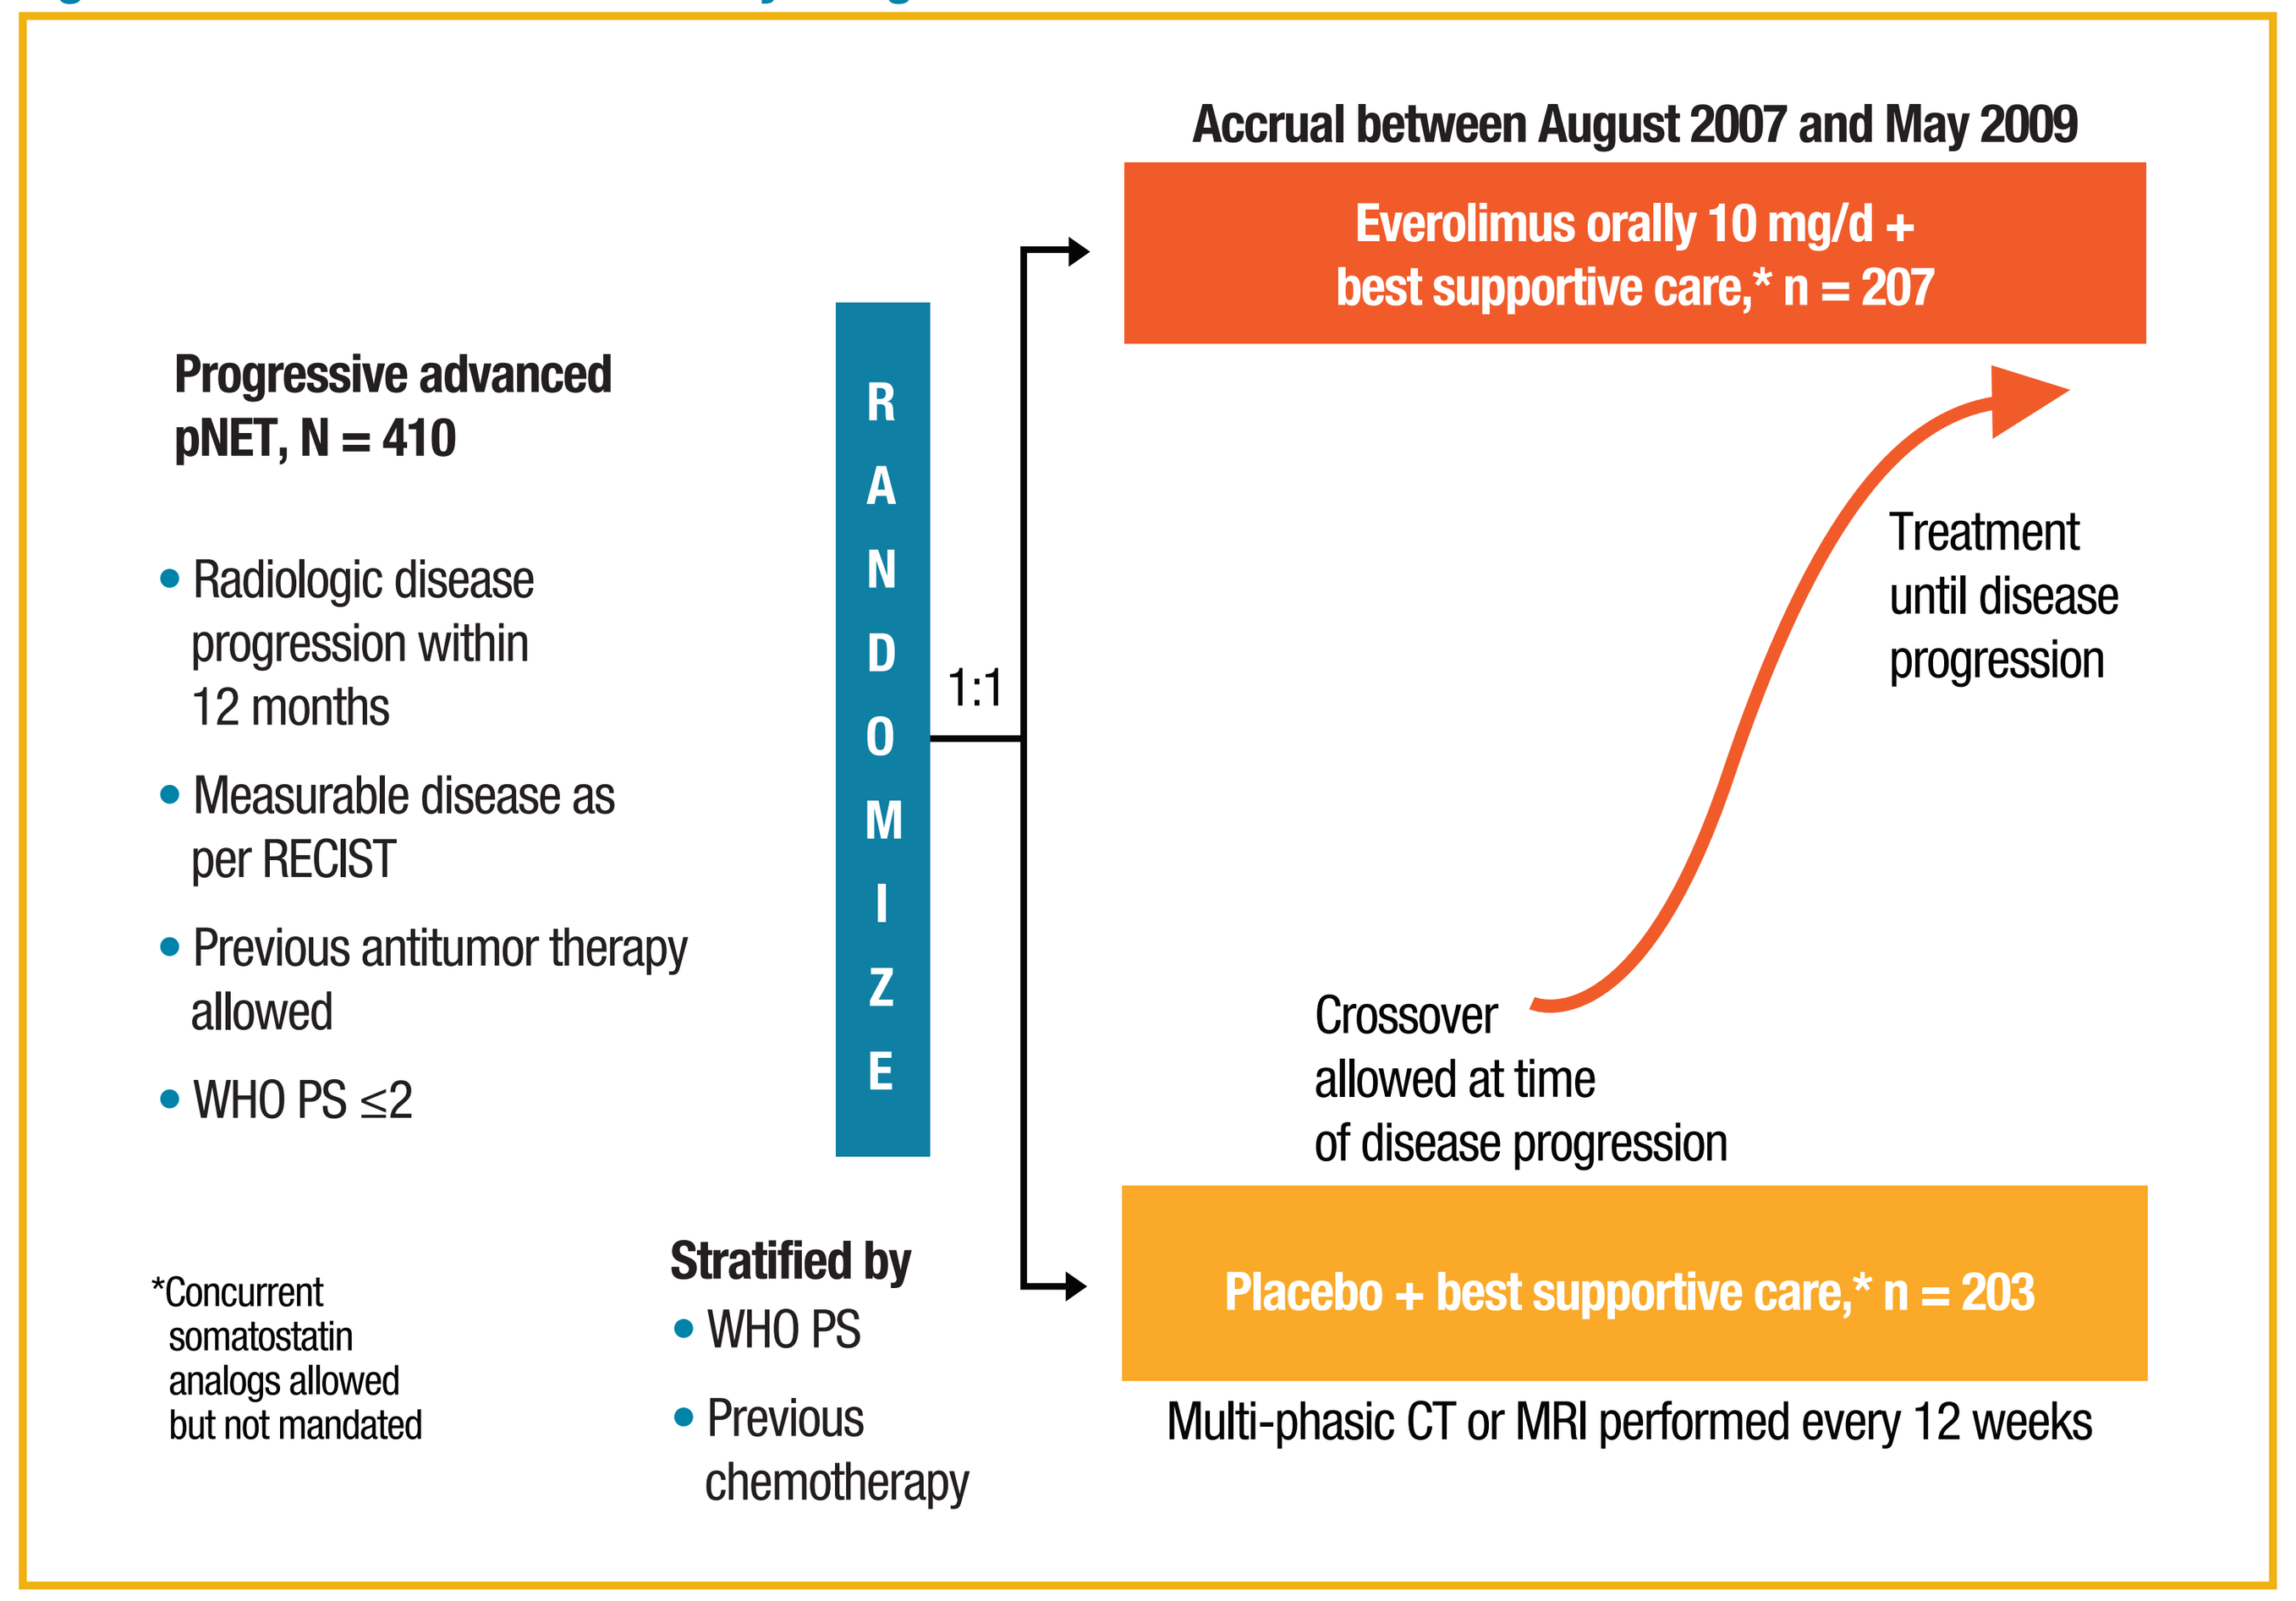
Accrual between August 2007 and May 2009
Everolimus orally 10 mg/d + best supportive care,* n = 207
Progressive advanced pNET, N = 410
• Radiologic disease progression within 12 months
• Measurable disease as per RECIST
• Previous antitumor therapy allowed
• WHO PS ≤2
R A N D O M I Z E
1:1
Treatment until disease progression
Crossover allowed at time of disease progression
*Concurrent somatostatin analogs allowed but not mandated
Stratified by
• WHO PS
• Previous chemotherapy
Placebo + best supportive care,* n = 203
Multi-phasic CT or MRI performed every 12 weeks

WHO PS, World Organization performance status.

- Crossover was allowed for patients randomly assigned to placebo at the time of disease progression
- Primary study endpoint was median PFS documented by the local investigator according to Response Evaluation Criteria In Solid Tumors (RECIST), version 1.0; secondary study endpoints included safety, tumor response, overall survival, and biomarker changes
- Adverse events (AEs) were collected and coded to a preferred term using the Medical Dictionary for Regulatory Activities and were assessed according to the National Cancer Institute Common Terminology Criteria for Adverse Events, version 3.0

RESULTS

Patient Demographics and Baseline Characteristics

- Baseline characteristics were well balanced between treatment arms (**Table 1**)

Table 1. Baseline Demographics and Disease Characteristics

	Everolimus n = 207	Placebo n = 203
Median age, y (range)	58 (23-87)	57 (20-82)
Sex, %		
Male	53	58
Female	47	42
Age group, %		
<65 y	71	75
≥65 y	29	25
Race, %		
Caucasian	75	82
Asian	19	17
Black	4	1
Other	1	1
WHO PS, %		
0	67	66
1	30	32
2	3	3
No. organs involved, %		
1	25	31
2	41	32
≥3	34	38
Histologic grade, %		
Well differentiated	82	84
Moderately differentiated	17	15
Previous therapy, %		
Systemic antitumor therapy	57	58
Chemotherapy	50	50
Targeted therapy	5	7
Immunotherapy	3	4
Hormone therapy	1	1
Other	10	13
Somatostatin analogs	49	50

WHO PS, World Health Organization performance status.
Cutoff date was February 28, 2010.

Patient Disposition

- Median treatment duration was 2.3 times longer for patients receiving everolimus than for those receiving placebo (37.8 weeks and 16.1 weeks, respectively)
- With the additional 52 weeks of follow-up, disease progression remained the primary reason for treatment discontinuation in both treatment arms

Overall Survival

- At the updated analysis cutoff date of February 23, 2011, median OS was 36.6 months in the placebo arm and had not been reached in the everolimus arm (HR, 0.89; 95% CI, 0.64-1.23) (**Figure 2**)
 - — In total, 68 deaths occurred in the everolimus arm and 78 deaths occurred in the placebo arm
- 172 of 203 patients (84.7%) initially randomly assigned to placebo crossed over to open-label everolimus at the time of disease progression
 - — These patients achieved a median OS of 36.6 months, which was likely the result of treatment with everolimus after crossover

Figure 2. Kaplan-Meier plot of overall survival.

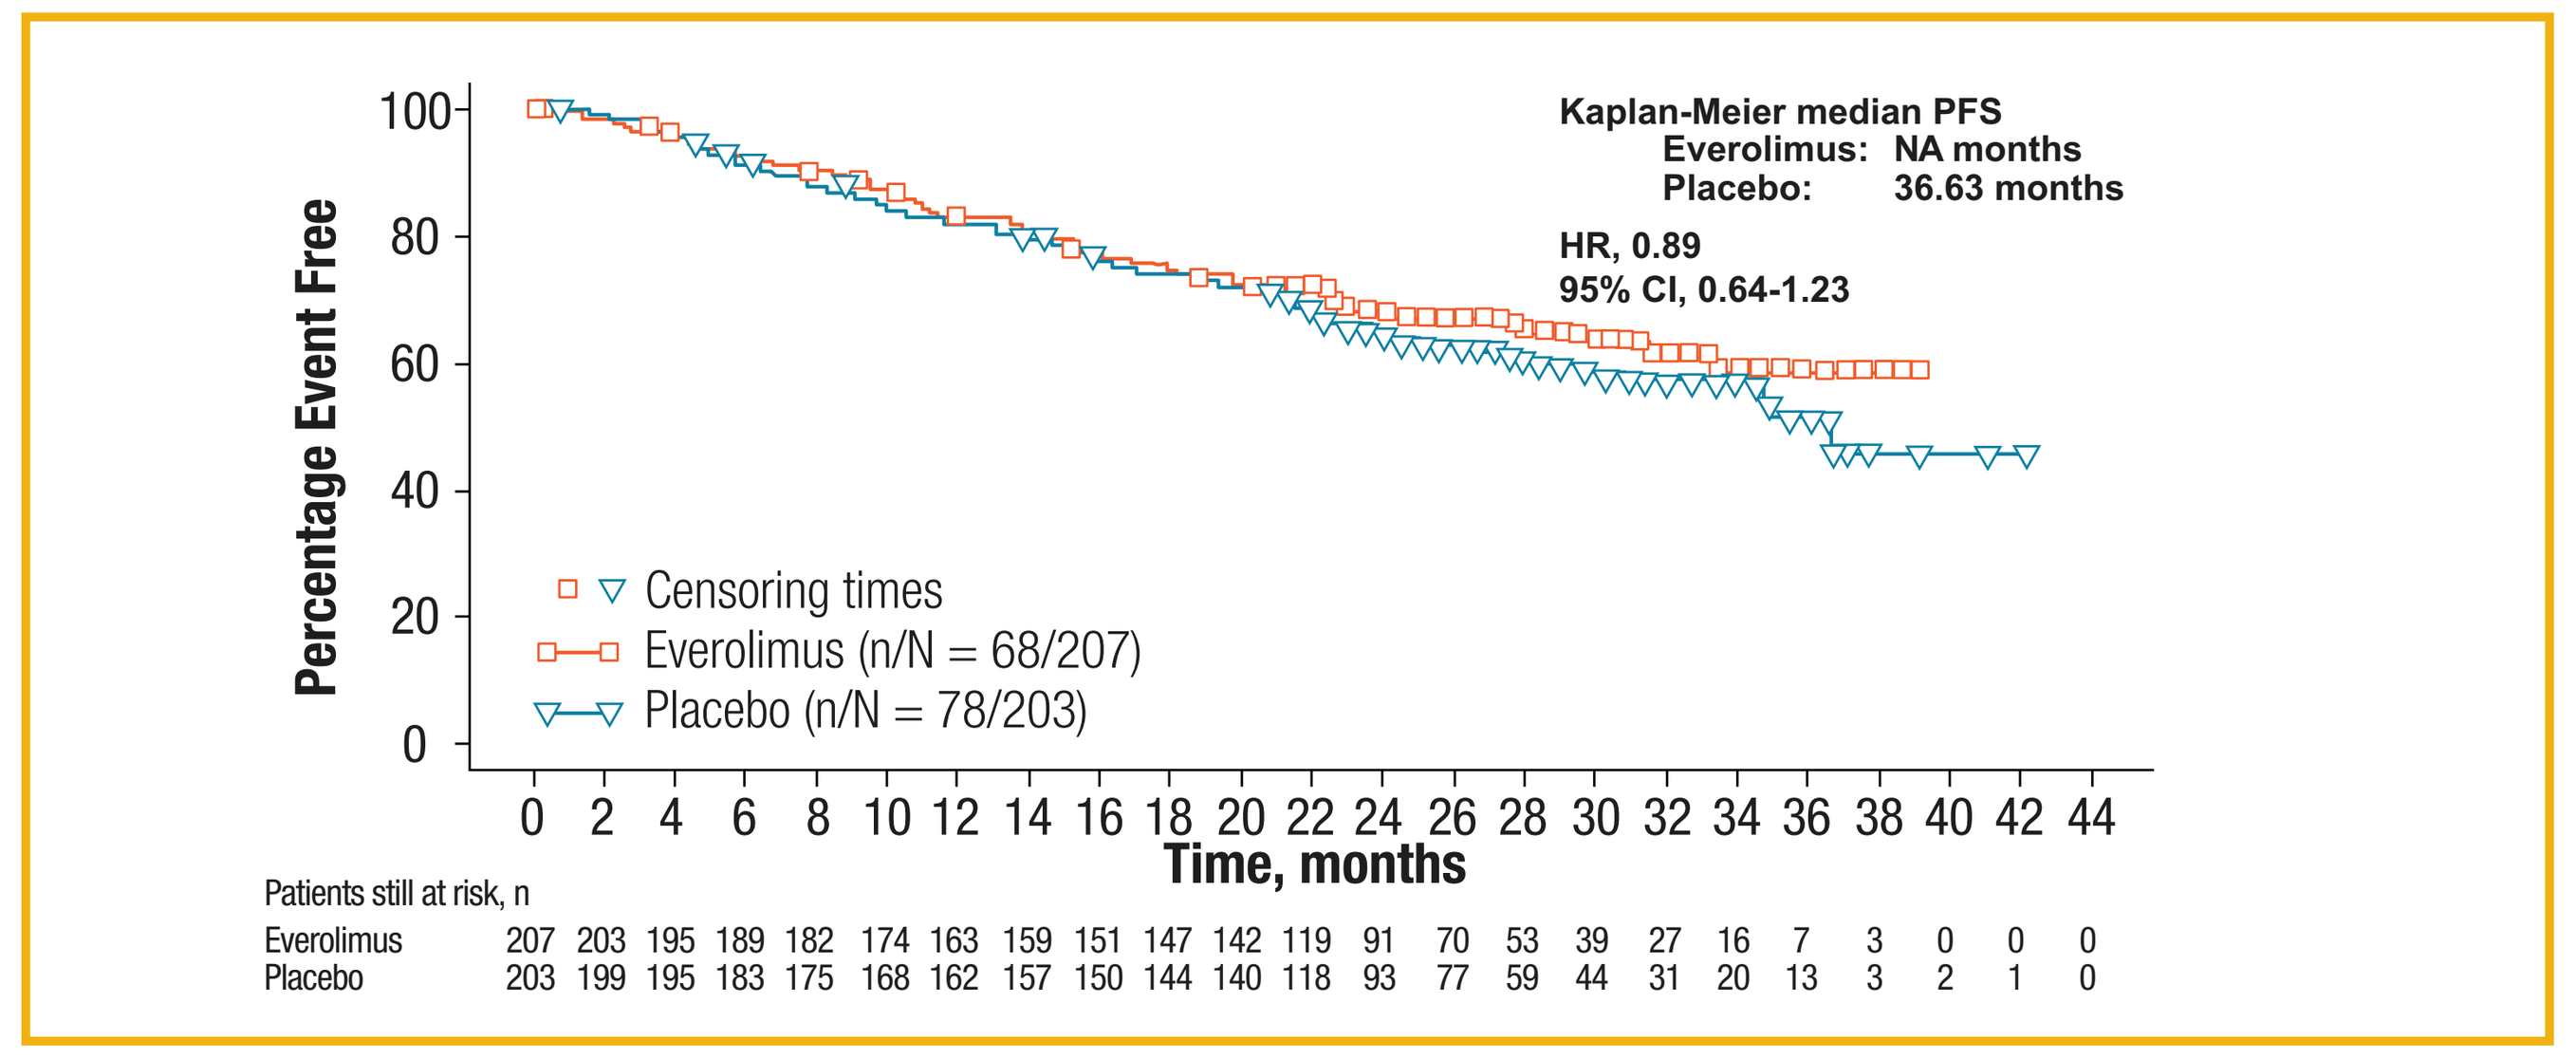
Kaplan-Meier median PFS
Everolimus: NA months
Placebo: 36.63 months
HR, 0.89
95% CI, 0.64-1.23
Percentage Event Free
100
80
60
40
20
0
Censoring times
Everolimus (n/N = 68/207)
Placebo (n/N = 78/203)
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30 32 34 36 38 40 42 44
Time, months
Patients still at risk, n
Everolimus 207 203 195 189 182 174 163 159 151 147 142 119 91 70 53 39 27 16 7 3 0 0 0
Placebo 203 199 195 183 175 168 162 157 150 144 140 118 93 77 59 44 31 20 13 3 2 1 0

Progression-Free Survival

- At the primary analysis cutoff date of February 28, 2010, median PFS was 11.0 months in patients receiving everolimus and 4.6 months in those receiving placebo (HR, 0.35; 95% CI, 0.27-0.45; *P* < 0.0001) (**Figure 3**)

Figure 3. Kaplan-Meier plot for PFS before crossover.

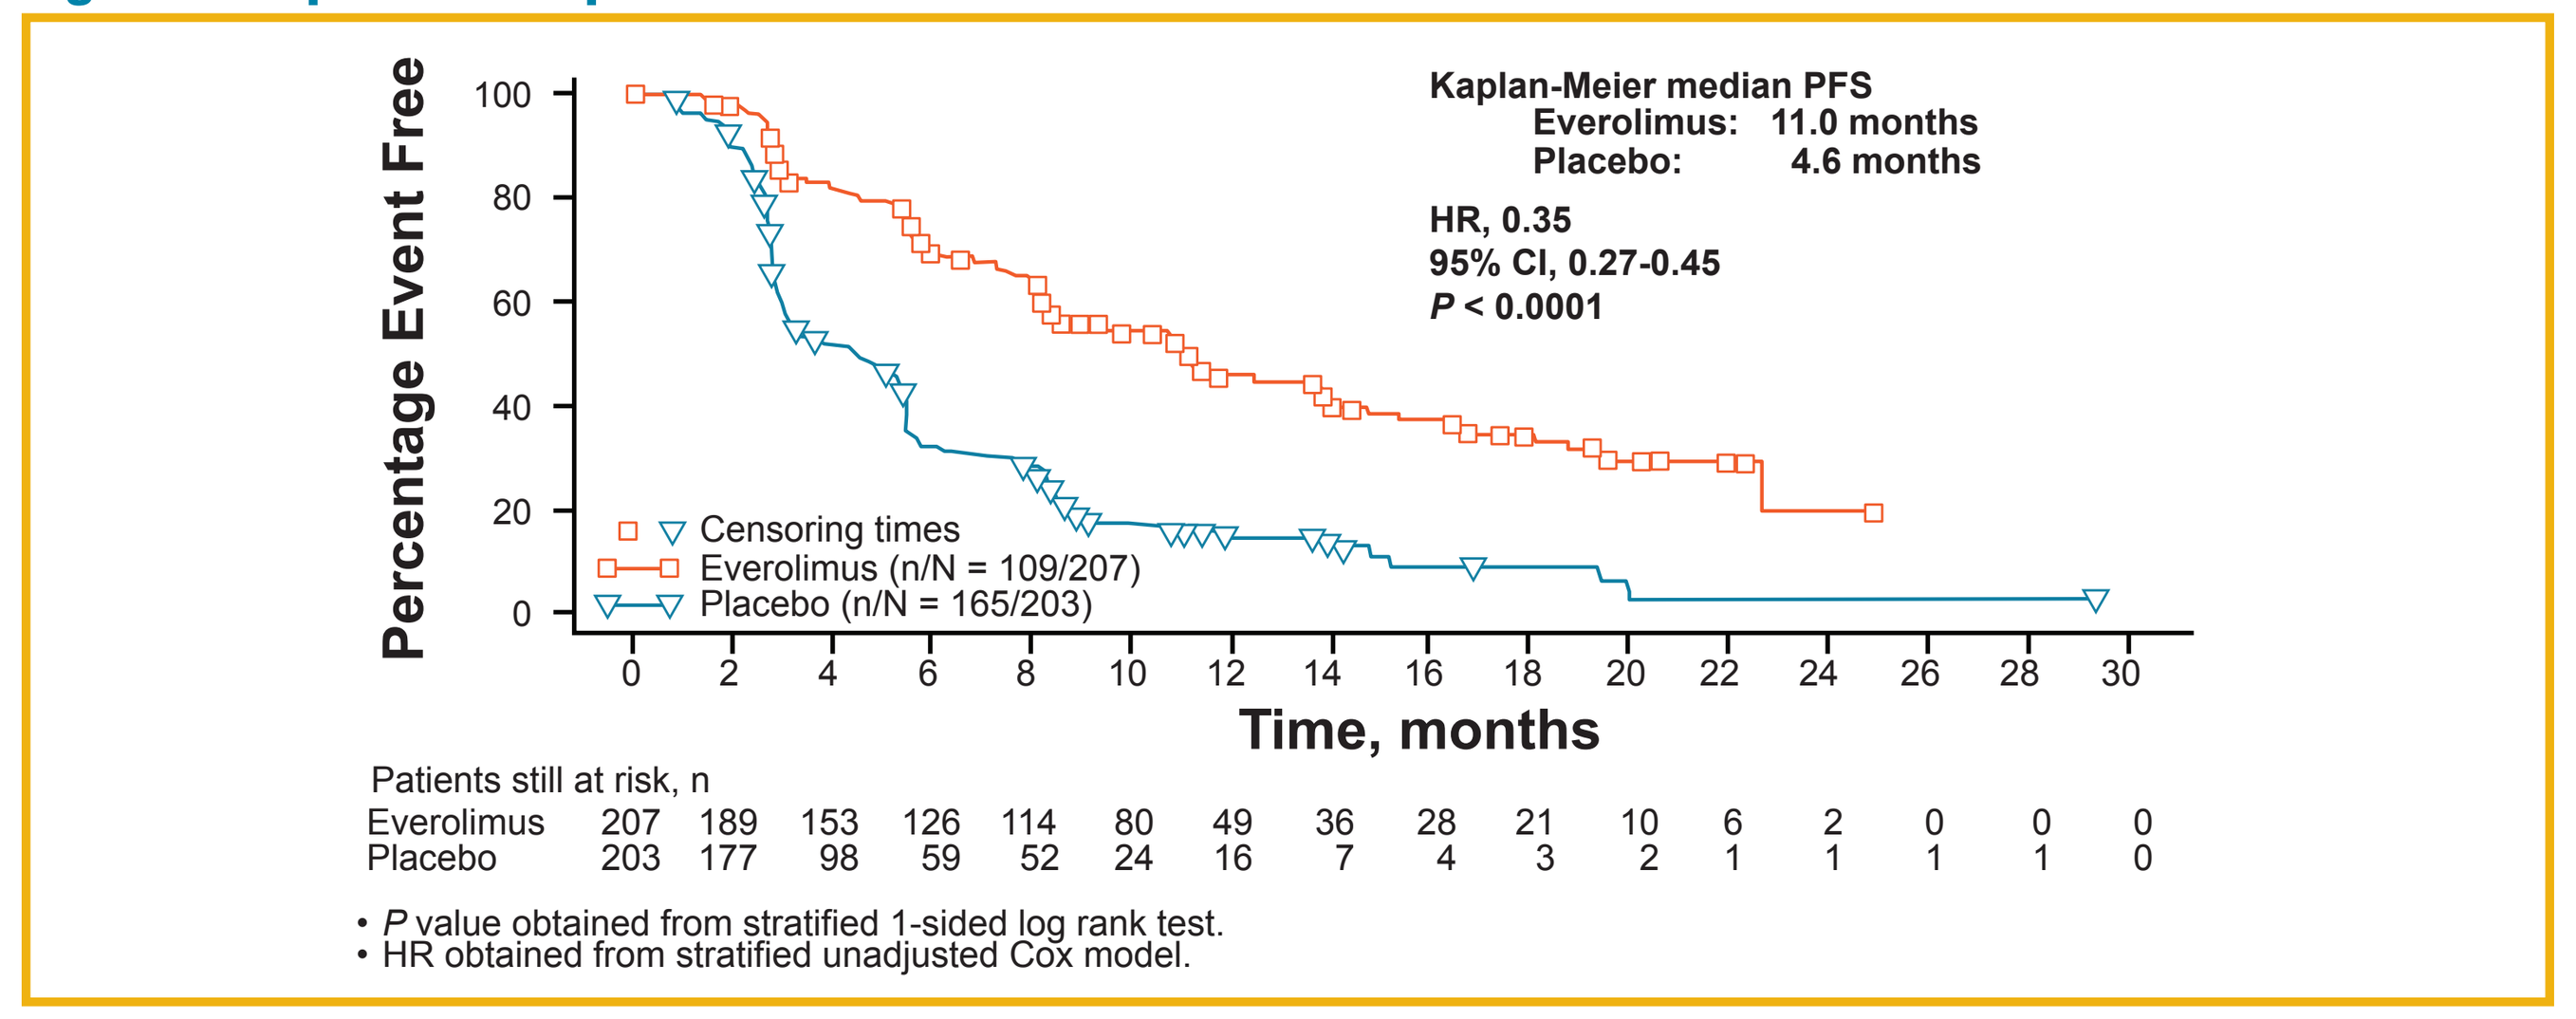
Kaplan-Meier median PFS
Everolimus: 11.0 months
Placebo: 4.6 months
HR, 0.35
95% CI, 0.27-0.45
P < 0.0001
Percentage Event Free
100
80
60
40
20
0
Censoring times
Everolimus (n/N = 109/207)
Placebo (n/N = 165/203)
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30
Time, months
Patients still at risk, n
Everolimus 207 189 153 126 114 80 49 36 28 21 10 6 2 0 0 0
Placebo 203 177 98 59 52 24 16 7 4 3 2 1 1 1 1 0
• P value obtained from stratified 1-sided log rank test.
• HR obtained from stratified unadjusted Cox model.

- At the cutoff date of February 28, 2010, median PFS for patients who received open-label everolimus after disease progression was 11.43 months (**Figure 4**)
 - — This was similar to the median PFS of 11.0 months in patients initially randomly assigned to everolimus; however, the fact that patients were initially assigned to placebo and were receiving everolimus after crossover likely confounded the results

Figure 4. Progression-free survival in placebo patients receiving open-label everolimus after disease progression.

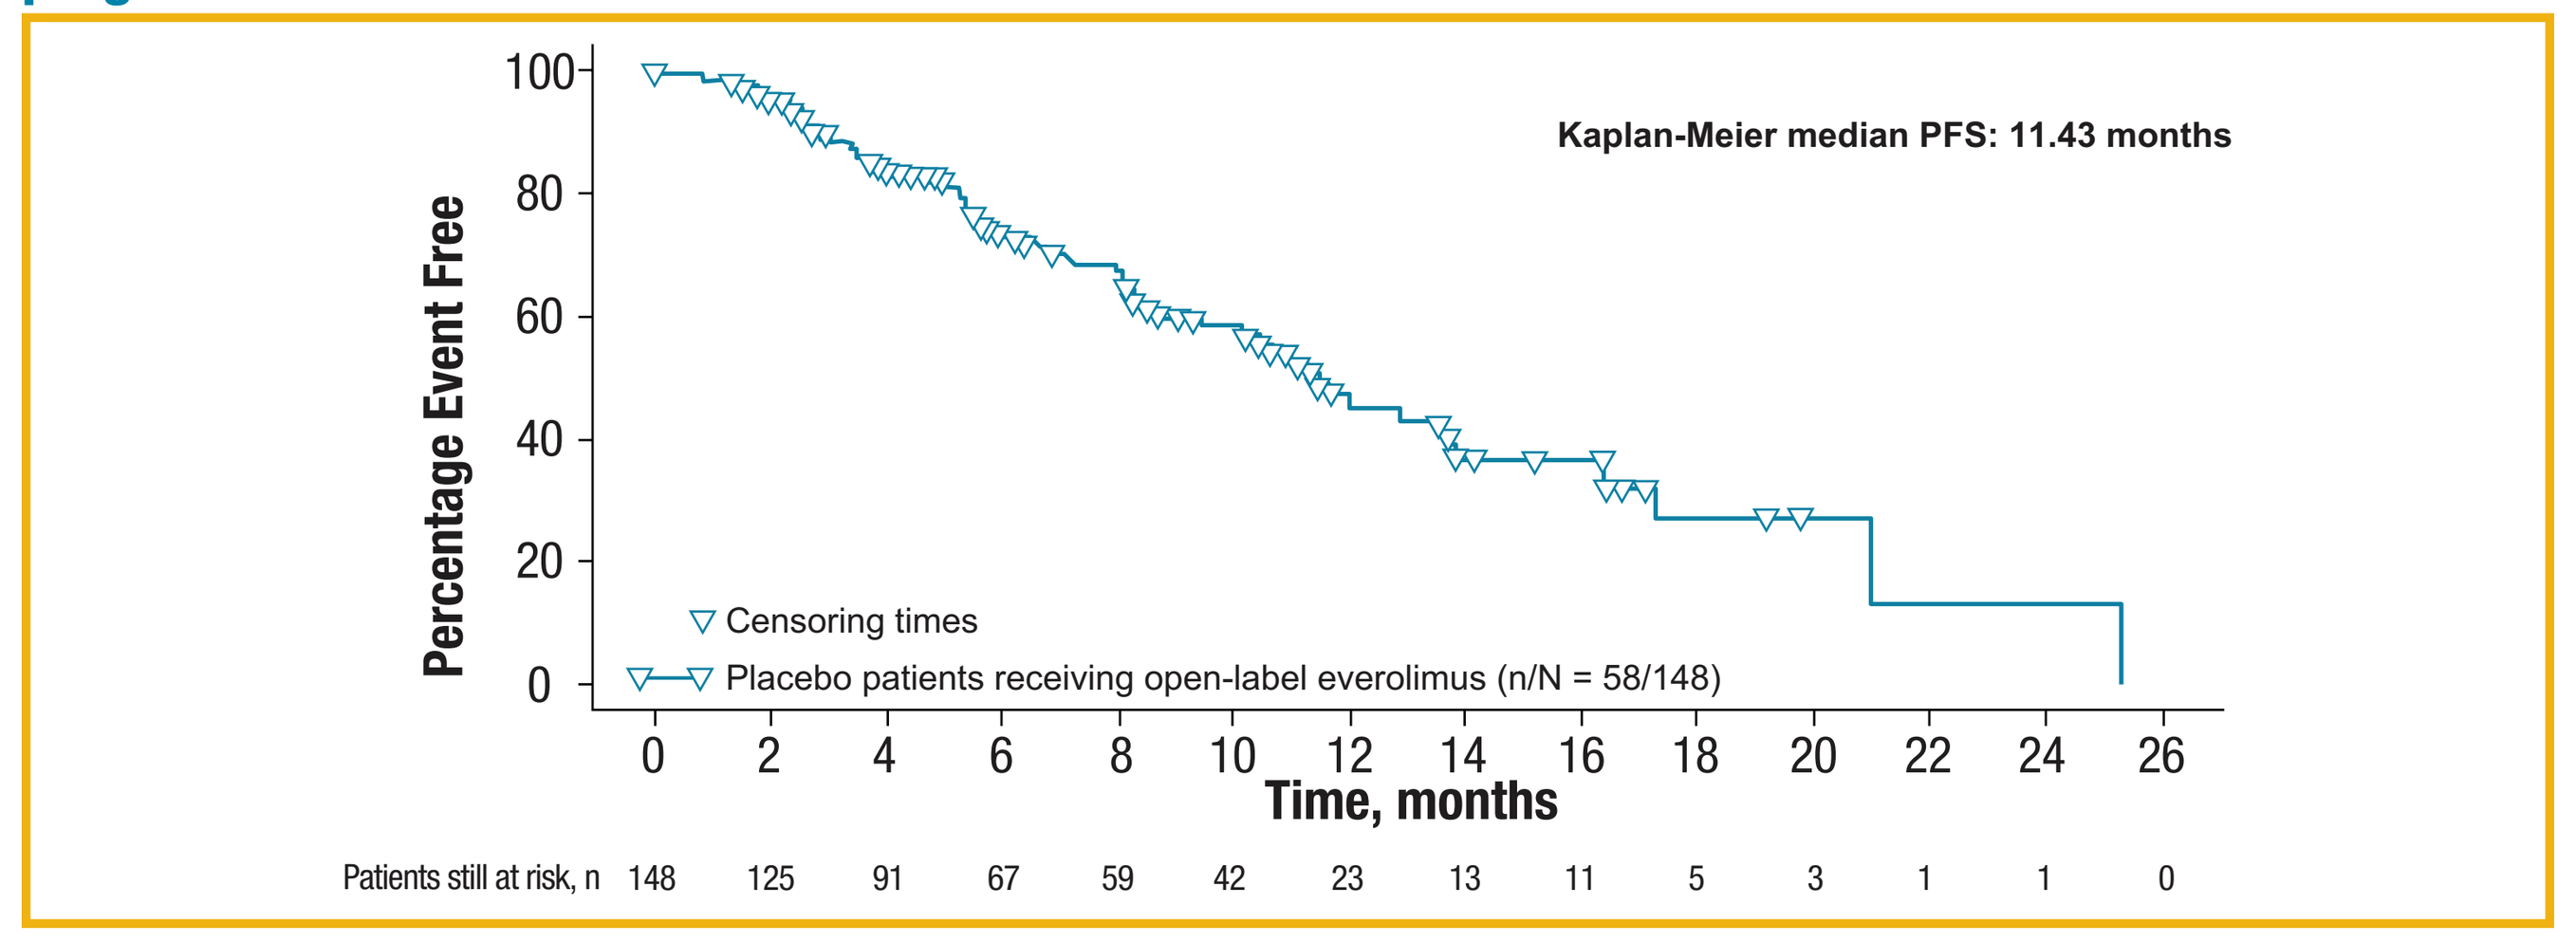
Kaplan-Meier median PFS: 11.43 months
Percentage Event Free
100
80
60
40
20
0
Censoring times
Placebo patients receiving open-label everolimus (n/N = 58/148)
0 2 4 6 8 10 12 14 16 18 20 22 24 26
Time, months
Patients still at risk, n 148 125 91 67 59 42 23 13 11 5 3 1 1 0

Safety

- At the 90-day safety analysis (cutoff date June 3, 2010), median follow-up was 20.1 months
- At the time of the extended safety analysis, 2.3-fold more patients allocated to everolimus remained on therapy compared with those allocated to placebo (24.0% and 10.3%, respectively)
- Treatment-related AE profiles remained unchanged in the 90-day safety analysis (**Table 2**)

Table 2. Treatment-Related Adverse Events Occurring in ≥5% of Patients

	Primary Analysis February 28, 2010		90-Day Safety Update June 3, 2010	
	Everolimus n = 204	Placebo n = 203	Everolimus n = 204	Placebo n = 203
Nonhematologic, %				
Stomatitis	52.9	11.3	52.9	12.3
Rash	48.5	10.3	48.5	10.3
Diarrhea	33.8	9.9	34.3	10.3
Fatigue	31.4	14.3	32.4	14.3
Infections and infestations	22.5	5.9	24.0	5.9
Peripheral edema	20.1	3.4	22.1	3.4
Headache	19.1	6.4	19.6	6.4
Nausea	20.1	18.2	20.1	18.2
Decreased appetite	19.6	6.9	20.1	6.9
Epistaxis	17.2	0	17.6	0
Dysgeusia	17.2	3.9	17.2	3.9
Decreased weight	15.7	4.4	16.7	4.4
Pulmonary events	16.7	0	16.7	0
Vomiting	15.2	6.4	15.2	6.4
Pruritus	14.7	8.9	14.7	8.9
Hyperglycemia	13.2	4.4	13.7	4.9
Asthenia	12.7	8.4	12.7	8.4
Nail disorder	11.8	1.0	12.7	1.0
Pyrexia	10.8	0	11.8	0
Cough	10.8	1.5	11.3	1.5
Aphthous stomatitis	10.8	3.4	10.8	3.4
Dry skin	10.3	4.4	10.3	4.4
Hypercholesterolemia	8.3	0	10.3	0
Hematologic, %				
Anemia	17.2	3.0	17.2	3.4
Thrombocytopenia	13.2	0.5	13.2	0.5
Neutropenia	6.4	2.0	6.4	2.0

- The most common nonhematologic treatment-related AEs with everolimus versus placebo were stomatitis (52.9% vs 12.3%), rash (48.5% vs 10.3%), diarrhea (34.3% vs 10.3%), and fatigue (32.4% vs 14.3%)
- The most common hematologic treatment-related AEs with everolimus versus placebo were anemia (17.2% vs 3.4%), thrombocytopenia (13.2% vs 0.5%), and neutropenia (6.4% vs 2.0%)
- During the extended safety follow-up, most frequently reported nonhematologic grade 3/4 AEs related to everolimus treatment remained hyperglycemia, stomatitis, and diarrhea (**Table 3**)
- Low rates of grade 3 hyperglycemia were seen with both everolimus and placebo (**Table 3**)
- No occurrences of grade 4 nonhematologic events were observed in the everolimus arm; grade 4 hyperglycemia was observed in 1% of patients in the placebo arm
- Most frequently reported treatment-related grade 3/4 hematologic AEs were anemia and thrombocytopenia (**Table 3**)
- Isolated treatment-related occurrences of grade 4 anemia (1.0%) and thrombocytopenia (<1%) were observed

Table 3. Treatment-Related Grade 3/4 Adverse Events Occurring in ≥3% of Patients

	Primary Analysis February 28, 2010		90-Day Safety Update June 3, 2010	
	Everolimus n = 204	Placebo n = 203	Everolimus n = 204	Placebo n = 203
Nonhematologic, %				
Hyperglycemia	5.4/0	1.5/0.5	5.9/0	1.5/1.0
Stomatitis	4.9/0	0/0	4.9/0	0/0
Diarrhea	3.4/0	0/0	3.4/0	0/0
Hematologic, %				
Anemia	4.9/1.0	0/0	4.9/1.0	0/0
Thrombocytopenia	3.4/0.5	0/0	3.4/0.5	0/0

- The most commonly reported AEs leading to the discontinuation of everolimus therapy were pneumonitis (3.4%), pyrexia (1.5%), fatigue, respiratory failure, interstitial lung disease, increased aspartate aminotransferase level, and pneumonia (1.0% each)
- The most commonly reported AEs leading to dose interruptions or reductions of everolimus therapy were stomatitis (10.3%), thrombocytopenia (7.8%), and pneumonitis (6.9%)

CONCLUSIONS

- **In the RADIANT-3 study—the largest randomized, double-blind, placebo-controlled, phase III trial in patients with advanced pNET—everolimus demonstrated a statistically and clinically significant 6-month improvement of median PFS (*P* < 0.0001; primary analysis cutoff date February 28, 2010)**
- **Results of the updated survival analysis (1 year after the primary analysis) show, at 40 months of follow-up, that median OS has not been reached in the everolimus arm; final analysis will be performed once approximately 250 deaths have occurred[10]**
- **Median OS of 36.6 months in the placebo arm, in which substantial crossover occurred benefiting these patients, exceeded the median OS of 24 months historically observed for patients with metastatic pNET[4]**
- **At a median follow-up of 20.1 months, updated safety results were consistent with those of the primary analysis and the known safety profile of everolimus in patients with cancer**
- **Most treatment-related AEs for everolimus were grade 1 or 2 and were manageable with proper planning, monitoring, and treatment**
- **No increase in AEs was observed with longer exposure to everolimus, demonstrating that the agent is tolerated long term**
- **These results support the use of everolimus as a standard treatment option for patients with progressing low- or intermediate-grade advanced pNET**

REFERENCES

1. Ries LAG et al, eds. Bethesda, MD: NIH Pub. No. 07-6215;2007.
2. Yao JC et al. *Ann Surg Oncol.* 2007;14:3492-3500.
3. Hauso O et al. *Cancer.* 2008;113:2655-2664.
4. Yao JC et al. *J Clin Oncol.* 2008;26:3063-3072.
5. Jensen RT et al. *Cancer.* 2008;113(suppl):1807-1843.
6. von Wichert G et al. *Cancer Res.* 2000;60:4573-4581.
7. Missiaglia E et al. *J Clin Oncol.* 2010;28:245-255.
8. Jiao Y et al. *Science.* 2011;331:1199-1203.
9. Yao JC et al. *J Clin Oncol.* 2010;28:69-76.
10. Yao JC et al. *N Engl J Med.* 2011;365:514-523.

The authors wish to acknowledge the contribution of Robert E. Winkler to this study.

BAR CODE TO BE PLACED HERE REMOVE BOX BORDER

http://XXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXX

Presented at the European Multidisciplinary Cancer Congress, 23–27 September 2011, Stockholm, Sweden

This study was sponsored by Novartis Pharmaceuticals Corporation.

BACKGROUND

- Pancreatic neuroendocrine tumors (pNET) account for approximately 1.3% of the incidence of all pancreatic cancers and 4% to 7% of the incidence of all NET, although both incidences are increasing[1-4]
- At diagnosis, nearly 65% of patients with pNET have advanced metastatic disease, which is associated with a median survival time of 24 months[4]
- Although the molecular pathogenesis of sporadic pNET is unknown, data suggest that the phosphatidylinositol 3-kinase/Akt/mammalian target of rapamycin (mTOR) pathway, which is important for the regulation of cell growth, proliferation, and angiogenesis, is involved[5-8]
- Everolimus, an oral mTOR inhibitor, has shown promising antitumor activity in a phase II study in patients with pNET[9]
- Everolimus was approved by the US Food and Drug Administration in May 2011 and was given a positive opinion by the European Union Committee for Medicinal Products for Human Use in July 2011 for the management of progressive pNET that is unresectable, locally advanced, or metastatic
- In the large, prospective, international, multicenter, randomized, double-blind, placebo-controlled, phase III RADIANT-3 trial involving 410 patients with progressing low- or intermediate-grade advanced pNET, everolimus 10 mg/d + best supportive care (BSC) compared with placebo + BSC demonstrated a statistically significant and clinically meaningful 6.4-month prolongation of median progression-free survival (PFS), as assessed by the local investigator (11.0 months vs 4.6 months; hazard ratio [HR], 0.35; 95% confidence interval [CI], 0.27-0.45; $P < 0.0001$)[10]
 - The benefit provided by everolimus was durable, as evident by the 34% of patients who remained free of disease progression at 18 months[10]

OBJECTIVE

- To evaluate an updated overall survival (OS) analysis (cutoff date February 23, 2011) and the 90-day safety update (cutoff date June 3, 2010) of everolimus + BSC versus placebo + BSC in patients who participated in the RADIANT-3 trial

METHODS

Study Design and Patient Population

- RADIANT-3 was an international, multicenter, double-blind, placebo-controlled, phase III study (**Figure 1**)

Figure 1. Phase III RADIANT-3 study design.

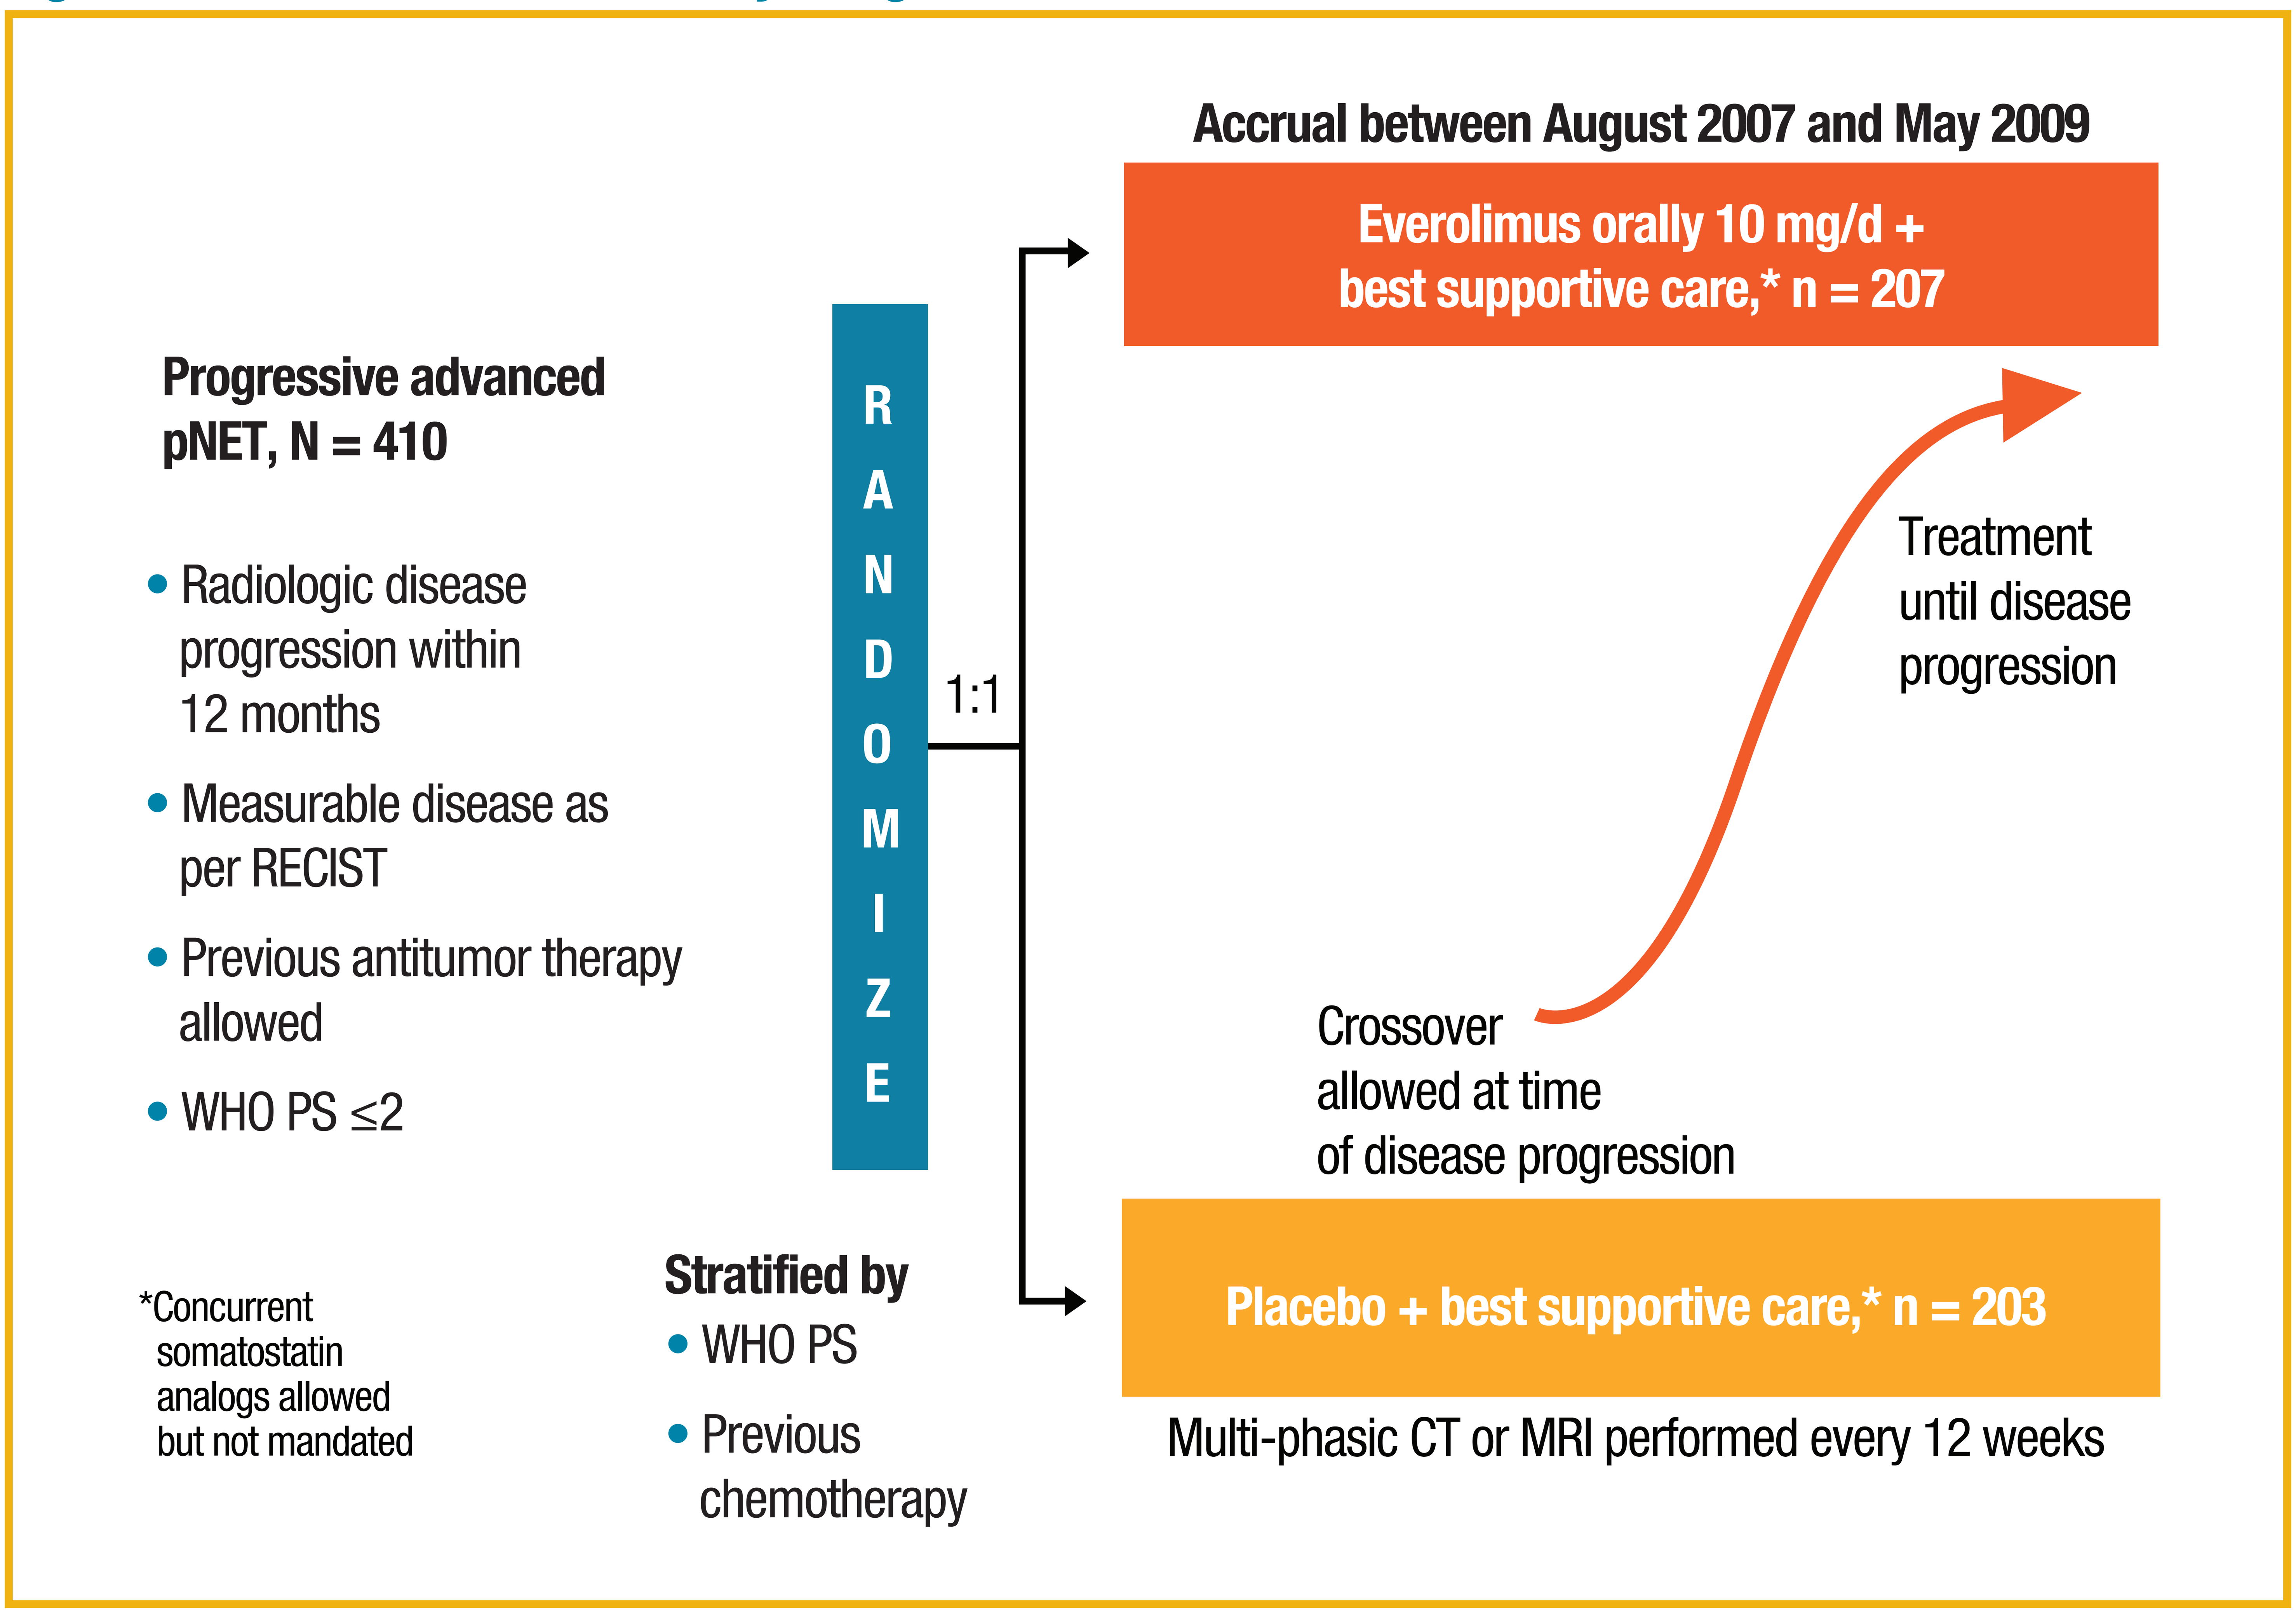

Accrual between August 2007 and May 2009
Everolimus orally 10 mg/d + best supportive care,* n = 207
Progressive advanced pNET, N = 410
Radiologic disease progression within 12 months
Measurable disease as per RECIST
Previous antitumor therapy allowed
WHO PS ≤2
RANDOMIZE
1:1
Treatment until disease progression
Crossover allowed at time of disease progression
Placebo + best supportive care,* n = 203
Multi-phasic CT or MRI performed every 12 weeks
*Concurrent somatostatin analogs allowed but not mandated
Stratified by
WHO PS
Previous chemotherapy

WHO PS, World Organization performance status.

- Crossover was allowed for patients randomly assigned to placebo at the time of disease progression
- Primary study endpoint was median PFS documented by the local investigator according to Response Evaluation Criteria In Solid Tumors (RECIST), version 1.0; secondary study endpoints included safety, tumor response, overall survival, and biomarker changes
- Adverse events (AEs) were collected and coded to a preferred term using the Medical Dictionary for Regulatory Activities and were assessed according to the National Cancer Institute Common Terminology Criteria for Adverse Events, version 3.0

Presented at the European Multidisciplinary Cancer Congress, 23–27 September 2011, Stockholm, Sweden

RESULTS

Patient Demographics and Baseline Characteristics

- Baseline characteristics were well balanced between treatment arms (**Table 1**)

Table 1. Baseline Demographics and Disease Characteristics

	Everolimus n = 207	Placebo n = 203
Median age, y (range)	58 (23-87)	57 (20-82)
Sex, %		
Male	53	58
Female	47	42
Age group, %		
<65 y	71	75
≥65 y	29	25
Race, %		
Caucasian	75	82
Asian	19	17
Black	4	1
Other	1	1
WHO PS, %		
0	67	66
1	30	32
2	3	3
No. organs involved, %		
1	25	31
2	41	32
≥3	34	38
Histologic grade, %		
Well differentiated	82	84
Moderately differentiated	17	15
Previous therapy, %		
Systemic antitumor therapy	57	58
Chemotherapy	50	50
Targeted therapy	5	7
Immunotherapy	3	4
Hormone therapy	1	1
Other	10	13
Somatostatin analogs	49	50

WHO PS, World Health Organization performance status.

Cutoff date was February 28, 2010.

Patient Disposition

- Median treatment duration was 2.3 times longer for patients receiving everolimus than for those receiving placebo (37.8 weeks and 16.1 weeks, respectively)
- With the additional 52 weeks of follow-up, disease progression remained the primary reason for treatment discontinuation in both treatment arms

Overall Survival

- At the updated analysis cutoff date of February 23, 2011, median OS was 36.6 months in the placebo arm and had not been reached in the everolimus arm (HR, 0.89; 95% CI, 0.64-1.23) (**Figure 2**)
 - In total, 68 deaths occurred in the everolimus arm and 78 deaths occurred in the placebo arm
- 172 of 203 patients (84.7%) initially randomly assigned to placebo crossed over to open-label everolimus at the time of disease progression
 - These patients achieved a median OS of 36.6 months, which was likely the result of treatment with everolimus after crossover

Figure 2. Kaplan-Meier plot of overall survival.

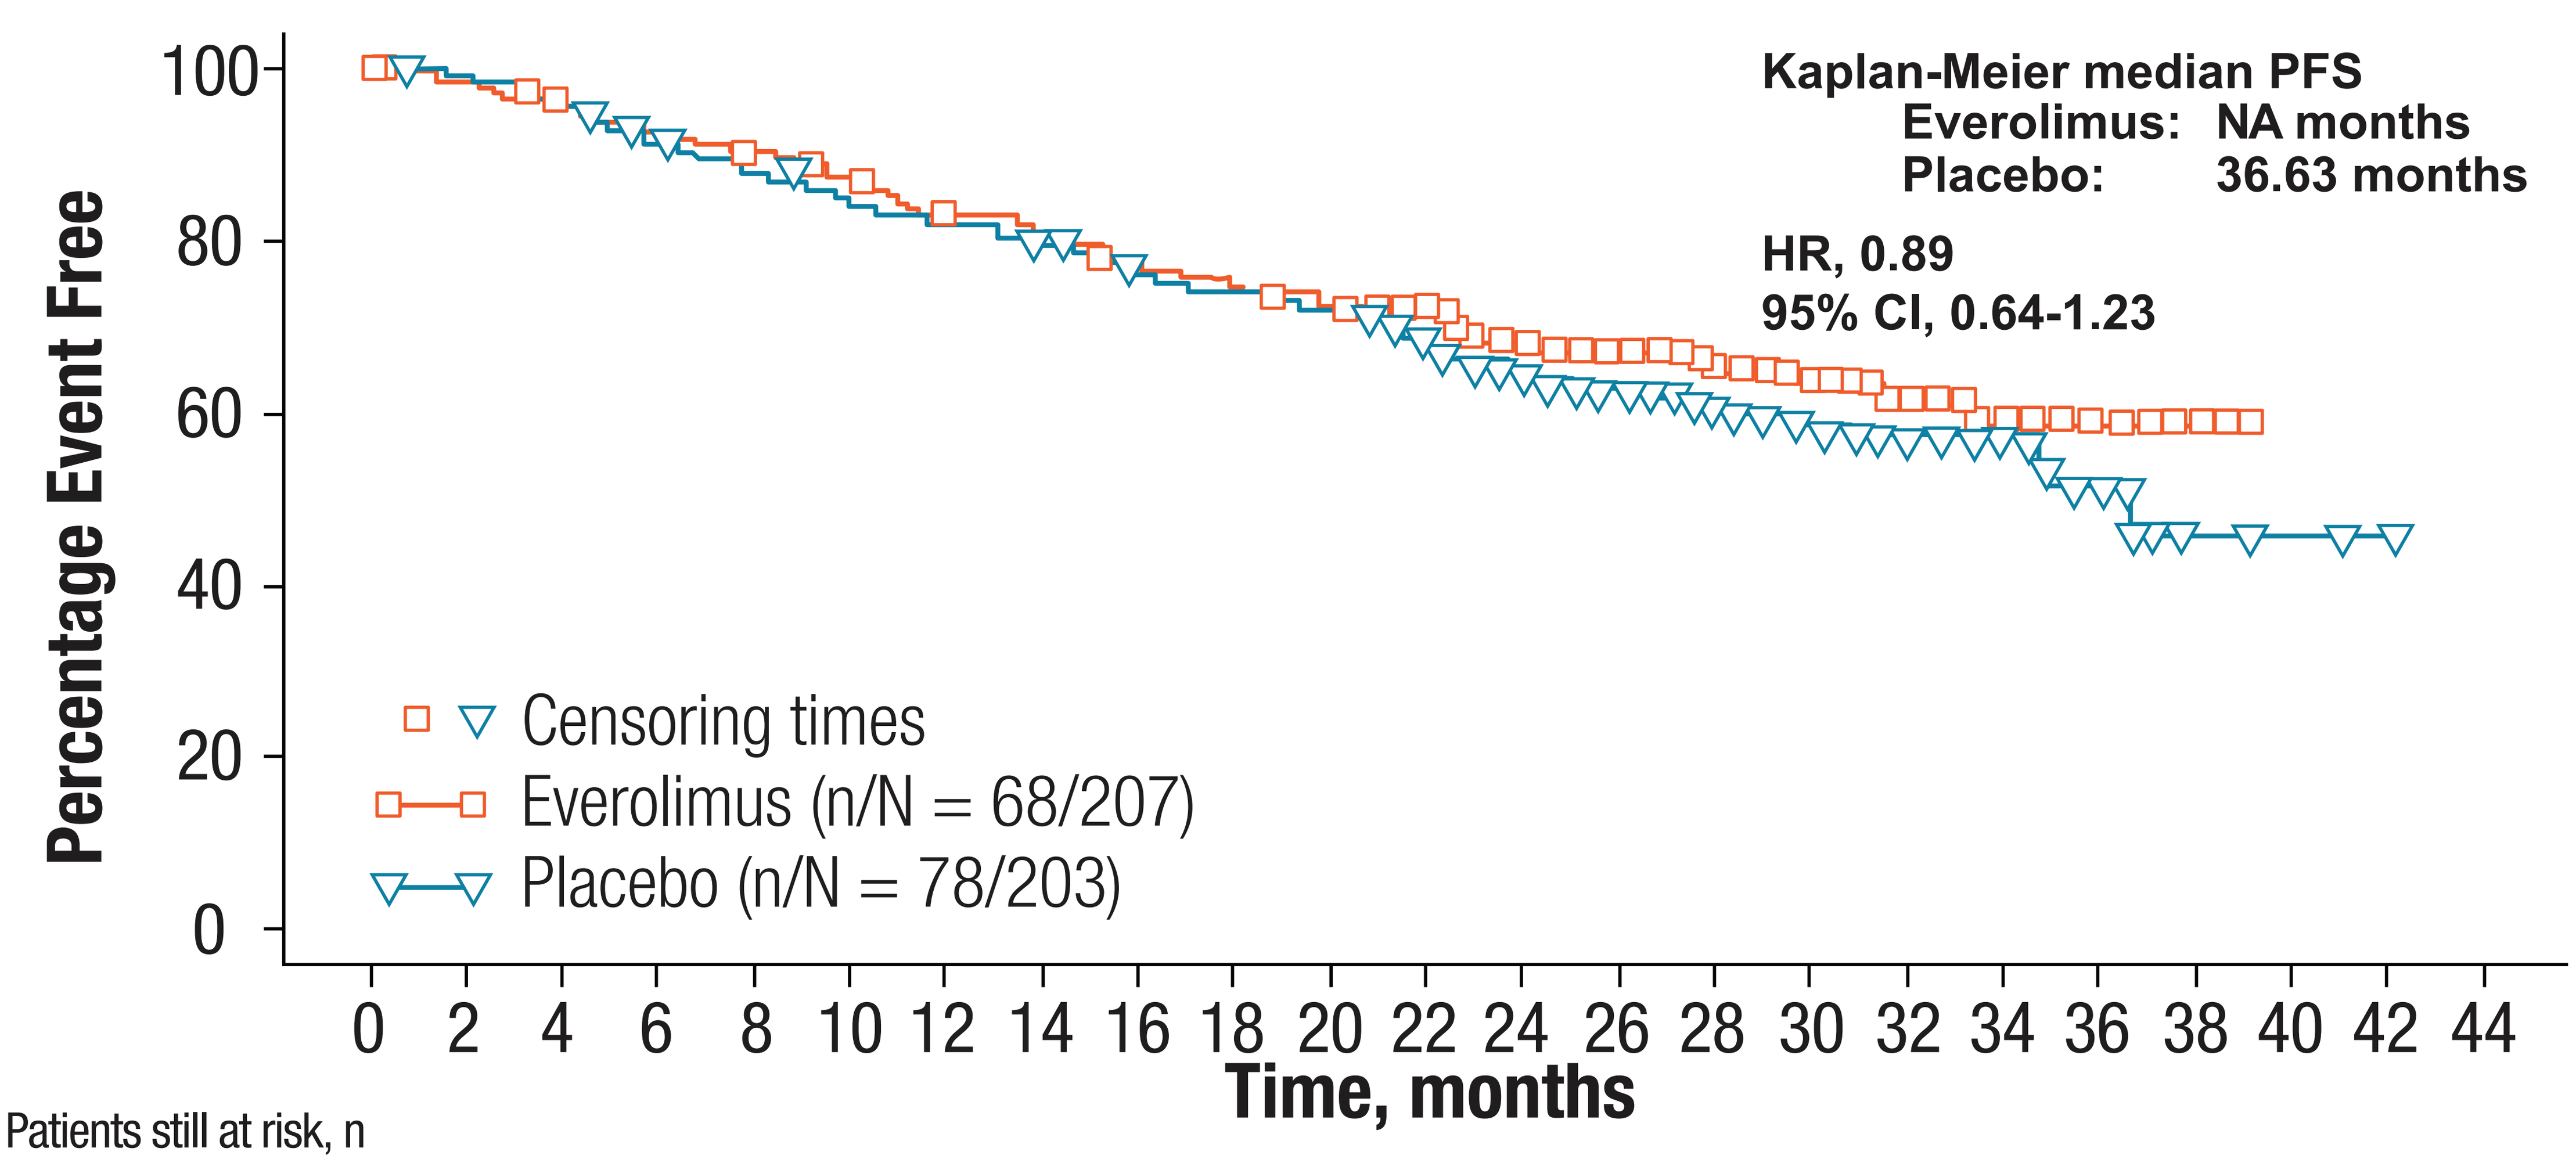

Kaplan-Meier median PFS
Everolimus: NA months
Placebo: 36.63 months
HR, 0.89
95% CI, 0.64-1.23
Percentage Event Free
100
80
60
40
20
0
Censoring times
Everolimus (n/N = 68/207)
Placebo (n/N = 78/203)
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30 32 34 36 38 40 42 44
Time, months

Patients still at risk, n

	0	2	4	6	8	10	12	14	16	18	20	22	24	26	28	30	32	34	36	38	40	42	
Everolimus	207	203	195	189	182	174	163	159	151	147	142	119	91	70	53	39	27	16	7	3	0	0	0
Placebo	203	199	195	183	175	168	162	157	150	144	140	118	93	77	59	44	31	20	13	3	2	1	0

Progression-Free Survival

- At the primary analysis cutoff date of February 28, 2010, median PFS was 11.0 months in patients receiving everolimus and 4.6 months in those receiving placebo (HR, 0.35; 95% CI, 0.27-0.45; *P* < 0.0001) (**Figure 3**)

Figure 3. Kaplan-Meier plot for PFS before crossover.

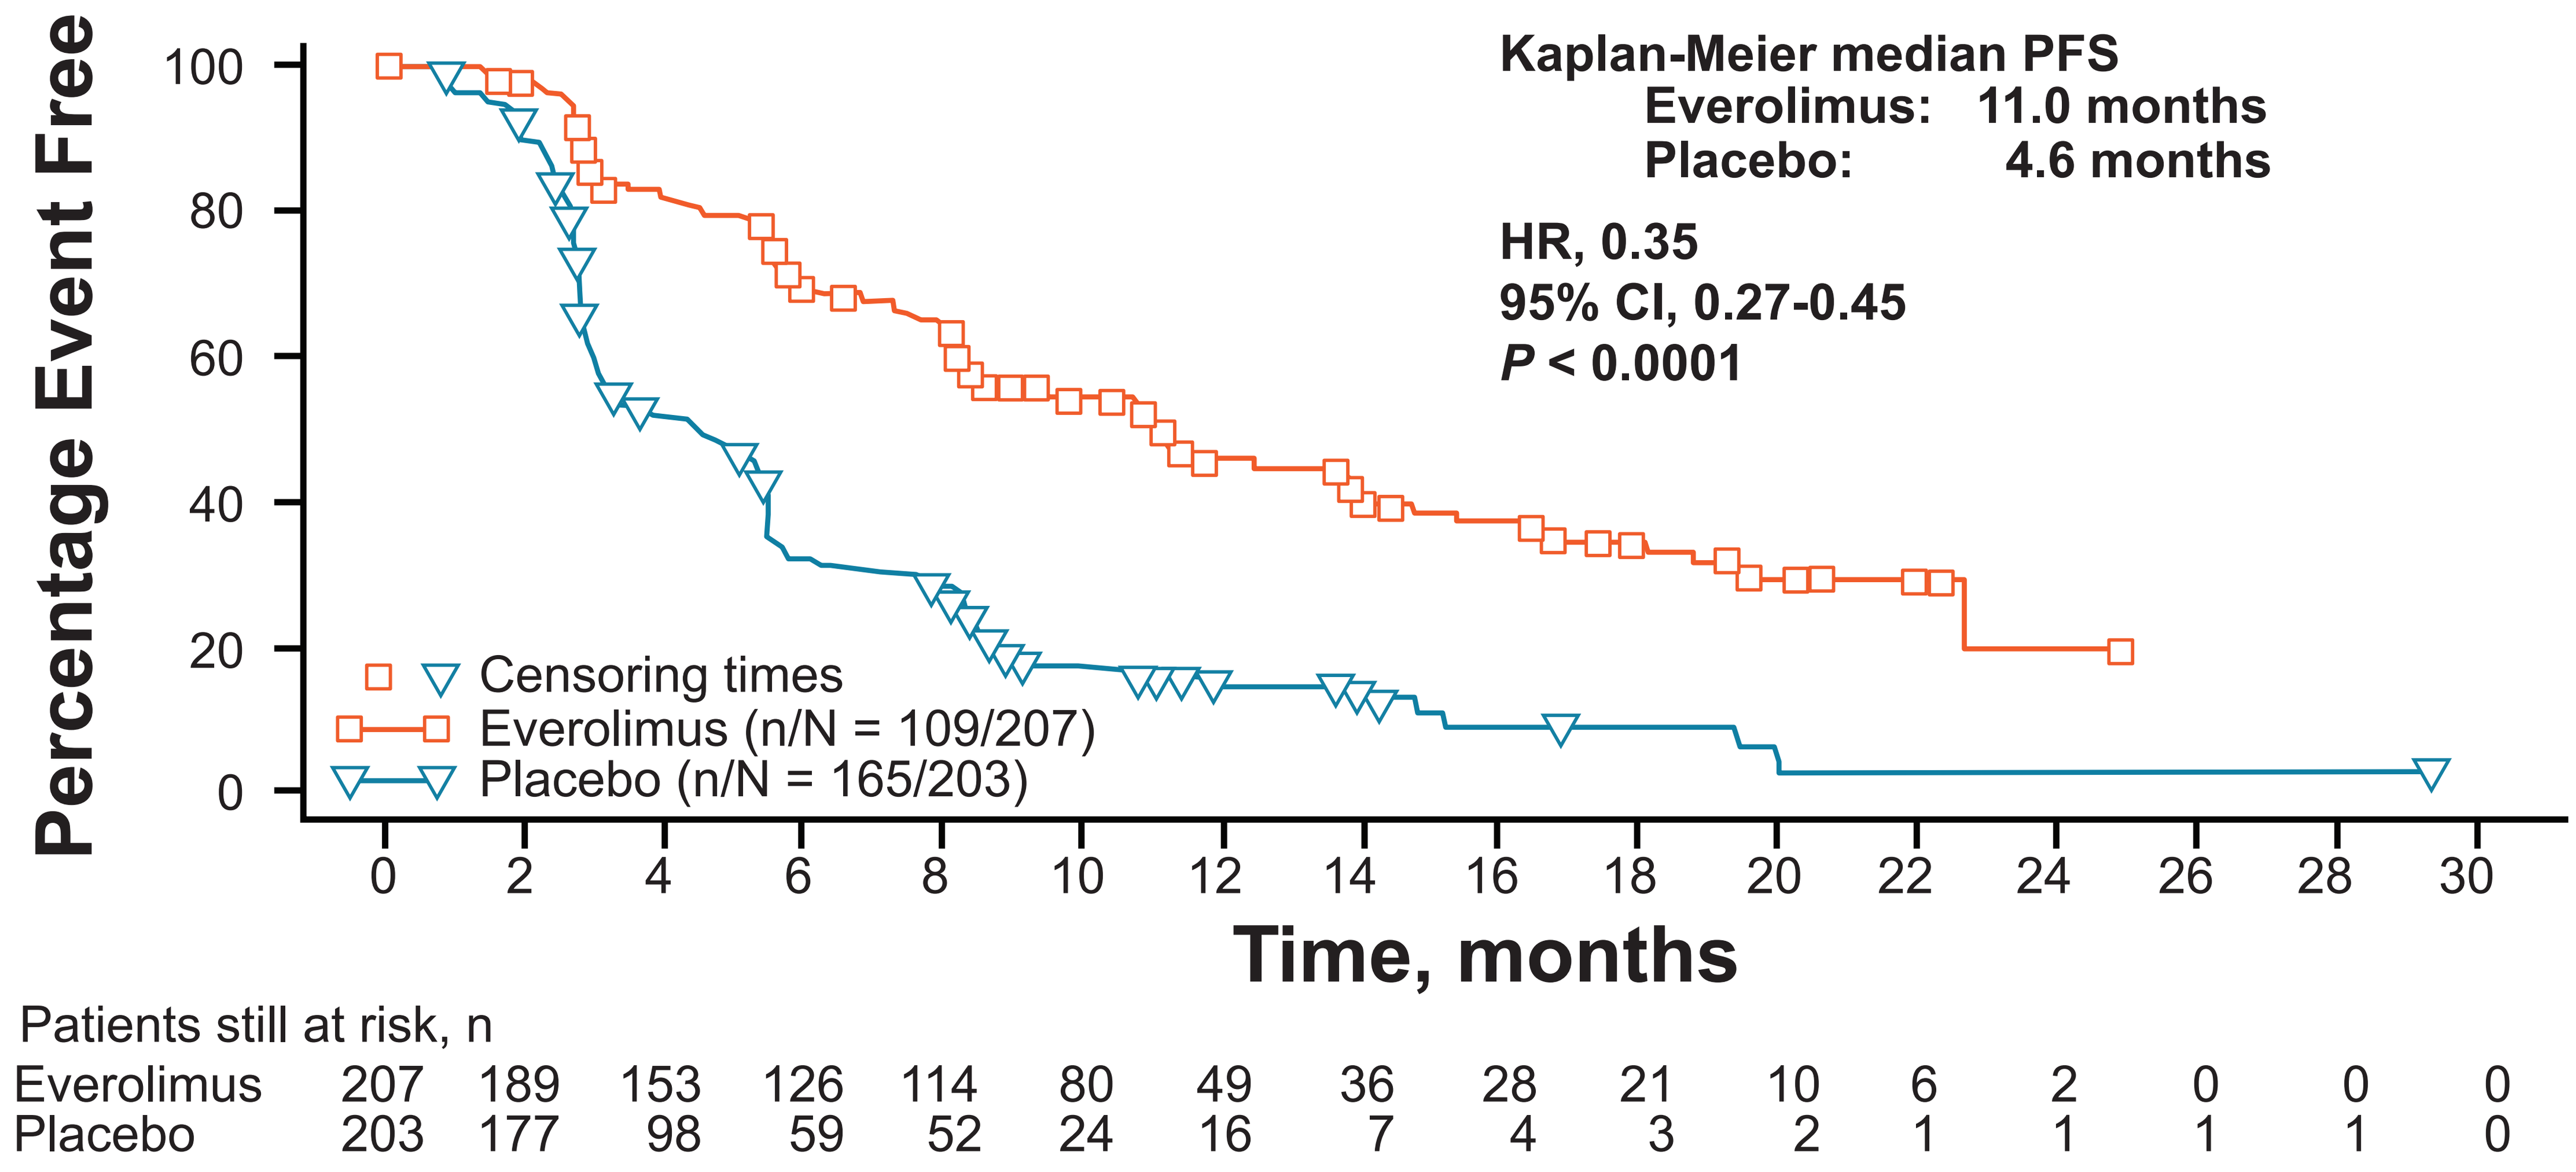
Kaplan-Meier median PFS
Everolimus: 11.0 months
Placebo: 4.6 months
HR, 0.35
95% CI, 0.27-0.45
P < 0.0001
Percentage Event Free
Censoring times
Everolimus (n/N = 109/207)
Placebo (n/N = 165/203)
Time, months
Patients still at risk, n
Everolimus 207 189 153 126 114 80 49 36 28 21 10 6 2 0 0 0
Placebo 203 177 98 59 52 24 16 7 4 3 2 1 1 1 1 0

- *P* value obtained from stratified 1-sided log rank test.
- HR obtained from stratified unadjusted Cox model.

- At the cutoff date of February 28, 2010, median PFS for patients who received open-label everolimus after disease progression was 11.43 months (**Figure 4**)
 - This was similar to the median PFS of 11.0 months in patients initially randomly assigned to everolimus; however, the fact that patients were initially assigned to placebo and were receiving everolimus after crossover likely confounded the results

Figure 4. Progression-free survival in placebo patients receiving open-label everolimus after disease progression.

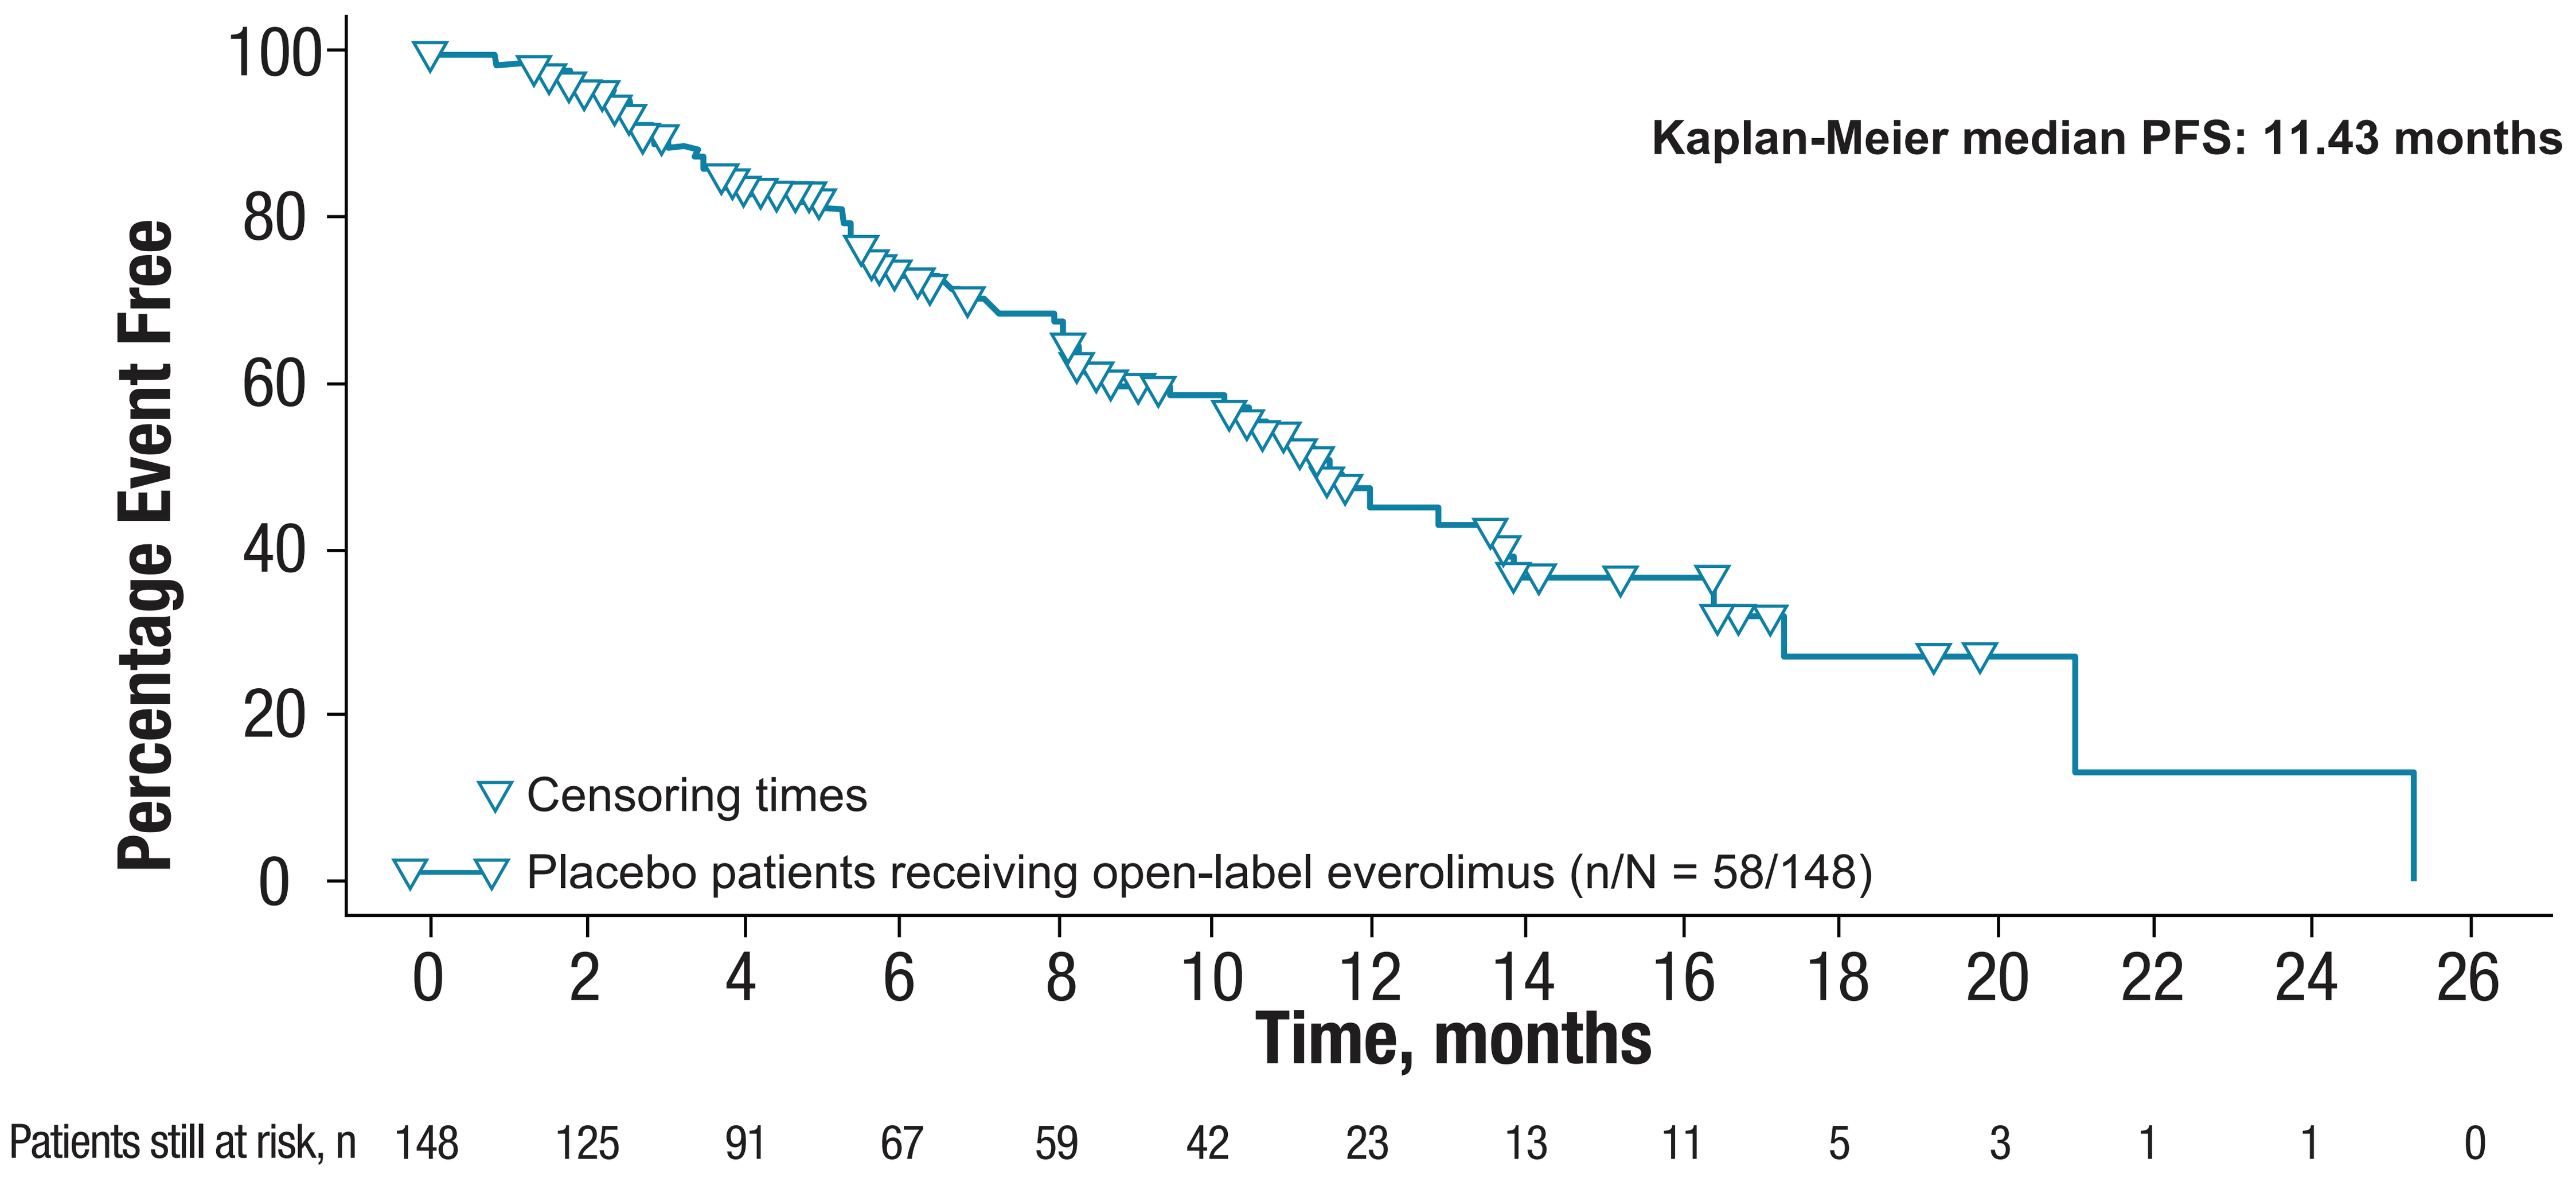
Kaplan-Meier median PFS: 11.43 months
Percentage Event Free
Censoring times
Placebo patients receiving open-label everolimus (n/N = 58/148)
Time, months
Patients still at risk, n 148 125 91 67 59 42 23 13 11 5 3 1 1 0

Safety

- At the 90-day safety analysis (cutoff date June 3, 2010), median follow-up was 20.1 months
- At the time of the extended safety analysis, 2.3-fold more patients allocated to everolimus remained on therapy compared with those allocated to placebo (24.0% and 10.3%, respectively)
- Treatment-related AE profiles remained unchanged in the 90-day safety analysis (**Table 2**)

Table 2. Treatment-Related Adverse Events Occurring in ≥5% of Patients

	Primary Analysis February 28, 2010		90-Day Safety Update June 3, 2010	
	Everolimus n = 204	Placebo n = 203	Everolimus n = 204	Placebo n = 203
Nonhematologic, %				
Stomatitis	52.9	11.3	52.9	12.3
Rash	48.5	10.3	48.5	10.3
Diarrhea	33.8	9.9	34.3	10.3
Fatigue	31.4	14.3	32.4	14.3
Infections and infestations	22.5	5.9	24.0	5.9
Peripheral edema	20.1	3.4	22.1	3.4
Headache	19.1	6.4	19.6	6.4
Nausea	20.1	18.2	20.1	18.2
Decreased appetite	19.6	6.9	20.1	6.9
Epistaxis	17.2	0	17.6	0
Dysgeusia	17.2	3.9	17.2	3.9
Decreased weight	15.7	4.4	16.7	4.4
Pulmonary events	16.7	0	16.7	0
Vomiting	15.2	6.4	15.2	6.4
Pruritus	14.7	8.9	14.7	8.9
Hyperglycemia	13.2	4.4	13.7	4.9
Asthenia	12.7	8.4	12.7	8.4
Nail disorder	11.8	1.0	12.7	1.0
Pyrexia	10.8	0	11.8	0
Cough	10.8	1.5	11.3	1.5
Aphthous stomatitis	10.8	3.4	10.8	3.4
Dry skin	10.3	4.4	10.3	4.4
Hypercholesterolemia	8.3	0	10.3	0
Hematologic, %				
Anemia	17.2	3.0	17.2	3.4
Thrombocytopenia	13.2	0.5	13.2	0.5
Neutropenia	6.4	2.0	6.4	2.0

- The most common nonhematologic treatment-related AEs with everolimus versus placebo were stomatitis (52.9% vs 12.3%), rash (48.5% vs 10.3%), diarrhea (34.3% vs 10.3%), and fatigue (32.4% vs 14.3%)
- The most common hematologic treatment-related AEs with everolimus versus placebo were anemia (17.2% vs 3.4%), thrombocytopenia (13.2% vs 0.5%), and neutropenia (6.4% vs 2.0%)
- During the extended safety follow-up, most frequently reported nonhematologic grade 3/4 AEs related to everolimus treatment remained hyperglycemia, stomatitis, and diarrhea (**Table 3**)
- Low rates of grade 3 hyperglycemia were seen with both everolimus and placebo (**Table 3**)
- No occurrences of grade 4 nonhematologic events were observed in the everolimus arm; grade 4 hyperglycemia was observed in 1% of patients in the placebo arm
- Most frequently reported treatment-related grade 3/4 hematologic AEs were anemia and thrombocytopenia (**Table 3**)
- Isolated treatment-related occurrences of grade 4 anemia (1.0%) and thrombocytopenia (<1%) were observed

Table 3. Treatment-Related Grade 3/4 Adverse Events Occurring in ≥3% of Patients

	Primary Analysis February 28, 2010		90-Day Safety Update June 3, 2010	
	Everolimus n = 204	Placebo n = 203	Everolimus n = 204	Placebo n = 203
Nonhematologic, %				
Hyperglycemia	5.4/0	1.5/0.5	5.9/0	1.5/1.0
Stomatitis	4.9/0	0/0	4.9/0	0/0
Diarrhea	3.4/0	0/0	3.4/0	0/0
Hematologic, %				
Anemia	4.9/1.0	0/0	4.9/1.0	0/0
Thrombocytopenia	3.4/0.5	0/0	3.4/0.5	0/0

- The most commonly reported AEs leading to the discontinuation of everolimus therapy were pneumonitis (3.4%), pyrexia (1.5%), fatigue, respiratory failure, interstitial lung disease, increased aspartate aminotransferase level, and pneumonia (1.0% each)
- The most commonly reported AEs leading to dose interruptions or reductions of everolimus therapy were stomatitis (10.3%), thrombocytopenia (7.8%), and pneumonitis (6.9%)

CONCLUSIONS

- **In the RADIANT-3 study—the largest randomized, double-blind, placebo-controlled, phase III trial in patients with advanced pNET—everolimus demonstrated a statistically and clinically significant 6-month improvement of median PFS (*P* < 0.0001; primary analysis cutoff date February 28, 2010)**
- **Results of the updated survival analysis (1 year after the primary analysis) show, at 40 months of follow-up, that median OS has not been reached in the everolimus arm; final analysis will be performed once approximately 250 deaths have occurred[10]**
- **Median OS of 36.6 months in the placebo arm, in which substantial crossover occurred benefiting these patients, exceeded the median OS of 24 months historically observed for patients with metastatic pNET[4]**
- **At a median follow-up of 20.1 months, updated safety results were consistent with those of the primary analysis and the known safety profile of everolimus in patients with cancer**
- **Most treatment-related AEs for everolimus were grade 1 or 2 and were manageable with proper planning, monitoring, and treatment**
- **No increase in AEs was observed with longer exposure to everolimus, demonstrating that the agent is tolerated long term**
- **These results support the use of everolimus as a standard treatment option for patients with progressing low- or intermediate-grade advanced pNET**

REFERENCES

1. Ries LAG et al, eds. Bethesda, MD: NIH Pub. No. 07-6215;2007.
2. Yao JC et al. *Ann Surg Oncol.* 2007;14:3492-3500.
3. Hauso O et al. *Cancer.* 2008;113:2655-2664.
4. Yao JC et al. *J Clin Oncol.* 2008;26:3063-3072.
5. Jensen RT et al. *Cancer.* 2008;113(suppl):1807-1843.
6. von Wichert G et al. *Cancer Res.* 2000;60:4573-4581.
7. Missiaglia E et al. *J Clin Oncol.* 2010;28:245-255.
8. Jiao Y et al. *Science.* 2011;331:1199-1203.
9. Yao JC et al. *J Clin Oncol.* 2010;28:69-76.
10. Yao JC et al. *N Engl J Med*. 2011;365:514-523.

The authors wish to acknowledge the contribution of Robert E. Winkler to this study.

BAR CODE TO BE PLACED HERE REMOVE BOX BORDER

http://XXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXXX

This study was sponsored by Novartis Pharmaceuticals Corporation.